Impact of Pegcetacoplan on Paroxysmal Nocturnal Hemoglobinuria Symptoms in Patients Previously Treated With C5 Inhibitors

Dharmik Desai, PharmD[1]; Nandini Hadker, MA[2]; Matthew O'Hara, MBA[2]; Laura Hill, BA[2]; Jed Perler, BA[2]; David Dingli, MD, PhD[3]

[1]Apellis Pharmaceuticals, Inc., Waltham, MA, USA; [2]Trinity Life Sciences, Waltham, MA, USA; [3]Division of Hematology, Department of Internal Medicine, Mayo Clinic, Rochester, MN, USA

OBJECTIVE

- To characterize the impact of pegcetacoplan compared with C5 inhibitors on the incidence, frequency, and severity of PNH symptoms in the real-world setting

CONCLUSIONS

- Patients experienced residual PNH symptoms with C5 inhibitors
- Symptom incidence, frequency, and severity improved after switching from C5 inhibitors to pegcetacoplan
- Evaluating patients' residual symptoms while on treatment is critical to optimize use of emerging treatments and improve HRQoL for patients with PNH

REFERENCES

1. Devos T, et al. *Eur J Haematol.* 2018;101(6):737–749.
2. Brodsky RA. *Blood.* 2014;124(18):2804–2811.
3. Bektas M, et al. *J Manag Care Spec Pharm.* 2020;26(12-b suppl):S8–S14.
4. Dingli D, et al. *Ann Hematol.* 2022;101(2):251–263.
5. Cella D, et al. *Ann Hematol.* 2022;101:1905–1914.
6. Ravulizumab-cwvz. Package insert. Alexion Pharmaceuticals, Inc; 2022.
7. Eculizumab. Package insert. Alexion Pharmaceuticals, Inc; 2022.
8. Pegcetacoplan. Package insert. Apellis Pharmaceuticals; 2021.
9. Pegcetacoplan. Summary of product characteristics. Swedish Orphan Biovitrum AB; 2022.
10. Hoy SM. *Drugs.* 2021;81(12):1423–1430.

Acknowledgments

This study was funded by Apellis Pharmaceuticals, Inc. Writing and editorial support were provided by Sarah Hauze, PhD (Kay Square Scientific, Newton Square, PA, USA). This support was funded by Apellis Pharmaceuticals, Inc., and Swedish Orphan Biovitrum AB. Apellis Pharmaceuticals and Swedish Orphan Biovitrum AB reviewed the poster.

Disclosures

Dharmik Desai: is an employee of and holds stock in Apellis Pharmaceuticals; **Nandini Hadker; Matthew O'Hara; Laura Hill;** and **Jed Perler:** nothing to disclose; **David Dingli:** consultancy for Alexion Pharmaceuticals, Apellis Pharmaceuticals, Bristol Myers Squibb, GlaxoSmithKline, Janssen Pharmaceuticals, Novartis, Takeda Pharmaceuticals, and Sanofi S.A.

Abbreviations

CV, cardiovascular; GI, gastrointestinal; HRQoL, health-related quality of life; PNH, paroxysmal nocturnal hemoglobinuria.

27th Annual International Congress on Hematologic Malignancies
February 23–26, 2023 | Miami Beach, FL | Virtual

CONTEXT

- Paroxysmal nocturnal hemoglobinuria (PNH) is a rare, acquired hematologic condition associated with significant morbidity, mortality, and impact on health-related quality of life (HRQoL)[1,2]
 - Patients experience the negative impact of PNH and related symptoms (including chronic anemia and fatigue) across physical, emotional, and social aspects of their lives[3,4]
- The PNH treatment paradigm has evolved with the introduction of complement component C5 inhibitors, including eculizumab and ravulizumab[5–7]
 - However, patients with PNH have reported considerable burden of illness while receiving these C5 inhibitors[4]
- Pegcetacoplan is the first targeted C3 therapy approved for adults with PNH (US) and adults with PNH plus anemia despite receiving C5-targeted therapy for ≥3 months (EU)[8–10]
- Patient experience using pegcetacoplan in the real-world has not been sufficiently documented
 - Since the burden of PNH persists despite available treatments, it is important to assess if there is any meaningful reduction in burden among patients switching from C5 inhibitors to pegcetacoplan

METHODS

Study design

- This exploratory mixed-methods, observational study enrolled US adult patients with PNH recruited from the Panther prescription database by a third-party recruiter
 - Study coordinators and sponsors were blinded to recruitment
- Consenting patients completed a screening questionnaire based on the inclusion/exclusion criteria
 - Eligible patients were ≥21 years of age with a physician-confirmed diagnosis of PNH for ≥6 months from study screening
 – Currently treated with and had ≥3 months of pegcetacoplan treatment
 – Willing to share experiences living with PNH in an interview

Data collection and analysis

- Qualitative interviews of approximately 60 minutes were conducted with eligible patients between February 2022–April 2022
- Three independent coders from Trinity Life Sciences implemented inductive coding of interview transcripts to capture emerging themes
- Interviews were grouped into key outcomes of interest including clinical burden, humanistic impact, HRQoL, and physical impact of PNH
 - Discrepancies among coders were resolved via an open discussion
- Patients scored symptoms using a 7-point Likert scale
 - Symptom severity ranged from 1 ("very mild") to 7 ("very severe")
 - Symptom frequency ranged from 1 ("never") to 7 ("always")
- Quantitative and aggregate data within each interview were reported using descriptive statistics (eg, means, medians, and range); no statistical hypothesis testing was conducted

RESULTS

Patient characteristics

- In total, 15 patients met inclusion criteria and completed the interviews
 - The median age was 40 years (range, 24.0–77.0 years) and 8 patients (53.3%) were female
 - All enrolled patients received treatment with a C5 inhibitor for a median duration of 24.0 months (range, 6.0–48.0 months)
 - Patients had a mean duration of pegcetacoplan treatment of 5.6 months (range, 3.0–10.0 months)
 - The mean time since PNH diagnosis in this cohort was 10 years (range, 1–30 years)

Burden of PNH symptoms

- A majority of patients (n=14; 93.3%) reported experiencing ≥1 PNH symptom while receiving C5 inhibitors (**Figure 1**)
 - Fatigue (n=14) was the most frequent symptom reported, followed by cognitive symptoms and shortness of breath (n=12 each), and muscle weakness and gastrointestinal (GI) symptoms (n=8 each)
- Patients provided these symptoms with a frequency rating (on a 7-point Likert scale) of 6.0 for fatigue, 5.4 each for cognitive symptoms and muscle weakness, and 5.3 for GI symptoms

Figure 1. Mean symptom prevalence by frequency and severity experienced on C5 inhibitors by patients interviewed (N=15)

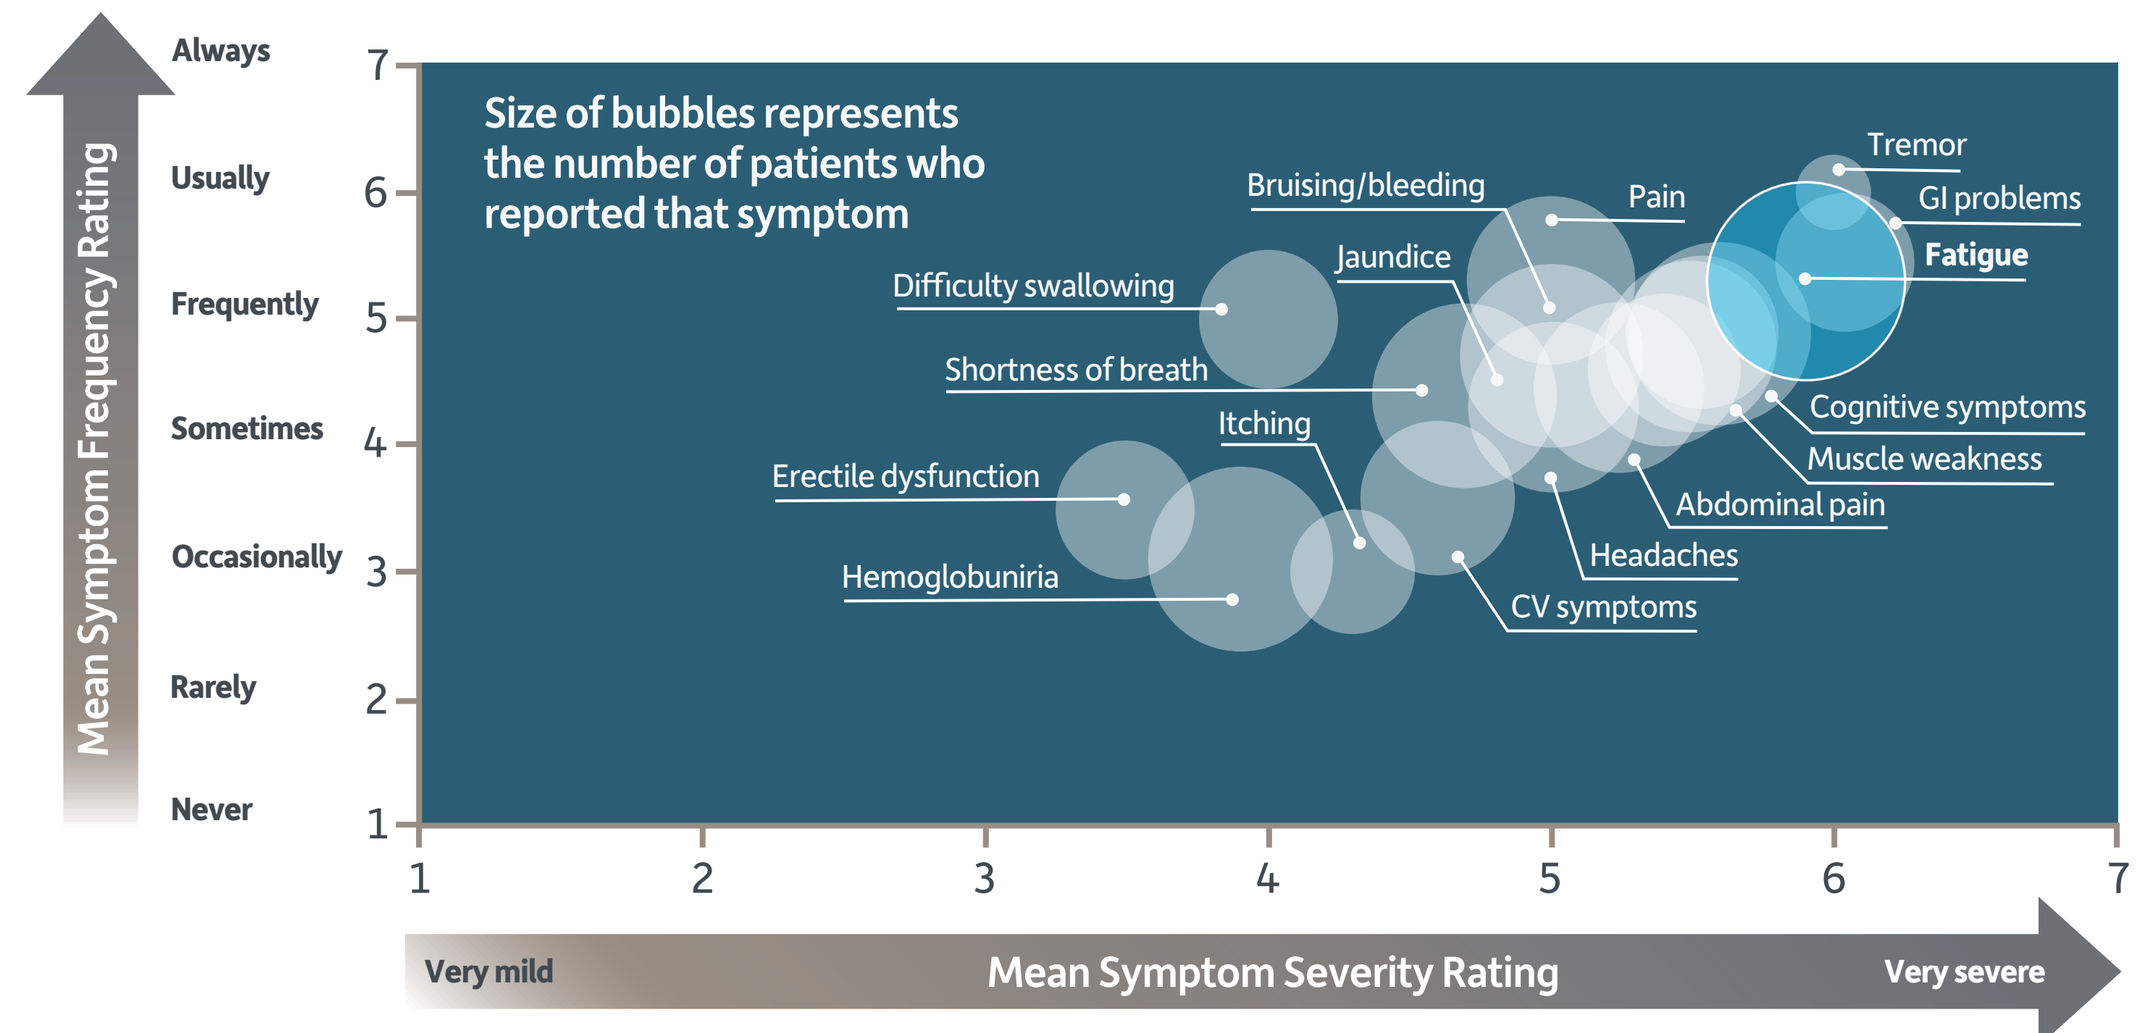
Always
Usually
Frequently
Sometimes
Occasionally
Rarely
Never
Mean Symptom Frequency Rating
Size of bubbles represents the number of patients who reported that symptom
Bruising/bleeding
Pain
Tremor
GI problems
Jaundice
Fatigue
Difficulty swallowing
Shortness of breath
Itching
Cognitive symptoms
Muscle weakness
Erectile dysfunction
Abdominal pain
Headaches
Hemoglobuniria
CV symptoms
Very mild
Mean Symptom Severity Rating
Very severe

Most frequent symptoms experienced with C5 inhibitors (mean rating)
Fatigue (6.0); Cognitive symptoms (5.4); Muscle weakness (5.4); GI symptoms (5.3)
Most severe symptoms experienced with C5 inhibitors (mean rating)
Fatigue (5.5); Pain (4.9); GI symptoms (4.9); Shortness of breath (4.8); Cognitive symptoms (4.8)
CV, cardiovascular; GI, gastrointestinal.

- Symptom severity was also moderate to high, with fatigue reported as a 5.5 (on a 7-point Likert scale), followed by pain and GI symptoms (each 4.9), and shortness of breath and cognitive symptoms (each 4.8)
- Just over half of patients (n=8; 53.3%) reported experiencing >10 symptoms while being treated with C5 inhibitors

Impact of switching to pegcetacoplan on PNH symptoms

- Eight patients (53.3%) reported an increase in average symptom-free days/month on pegcetacoplan (mean 13.1 days/month) compared with C5 inhibitors (mean 6.4 days/month)
- Symptoms of PNH reported by fewer patients on pegcetacoplan vs C5 inhibitors included:
 - Fatigue (n=12 vs n=14)
 - Cognitive symptoms (n=11 vs n=12)
 - Shortness of breath (n=7 vs n=12)
 - Pain (n=7 vs n=10)
 - Cardiovascular (CV) symptoms (n=6 vs n=11)
 - Muscle weakness (n=5 vs n=8)
 - GI symptoms (n=5 vs n=8)
- Patients reported a reduction in symptom frequency and severity after switching from C5 inhibitors to pegcetacoplan (**Figure 2**)

Figure 2. Mean symptom prevalence by frequency and severity experienced on pegcetacoplan versus C5 inhibitors by patients interviewed (n=15)

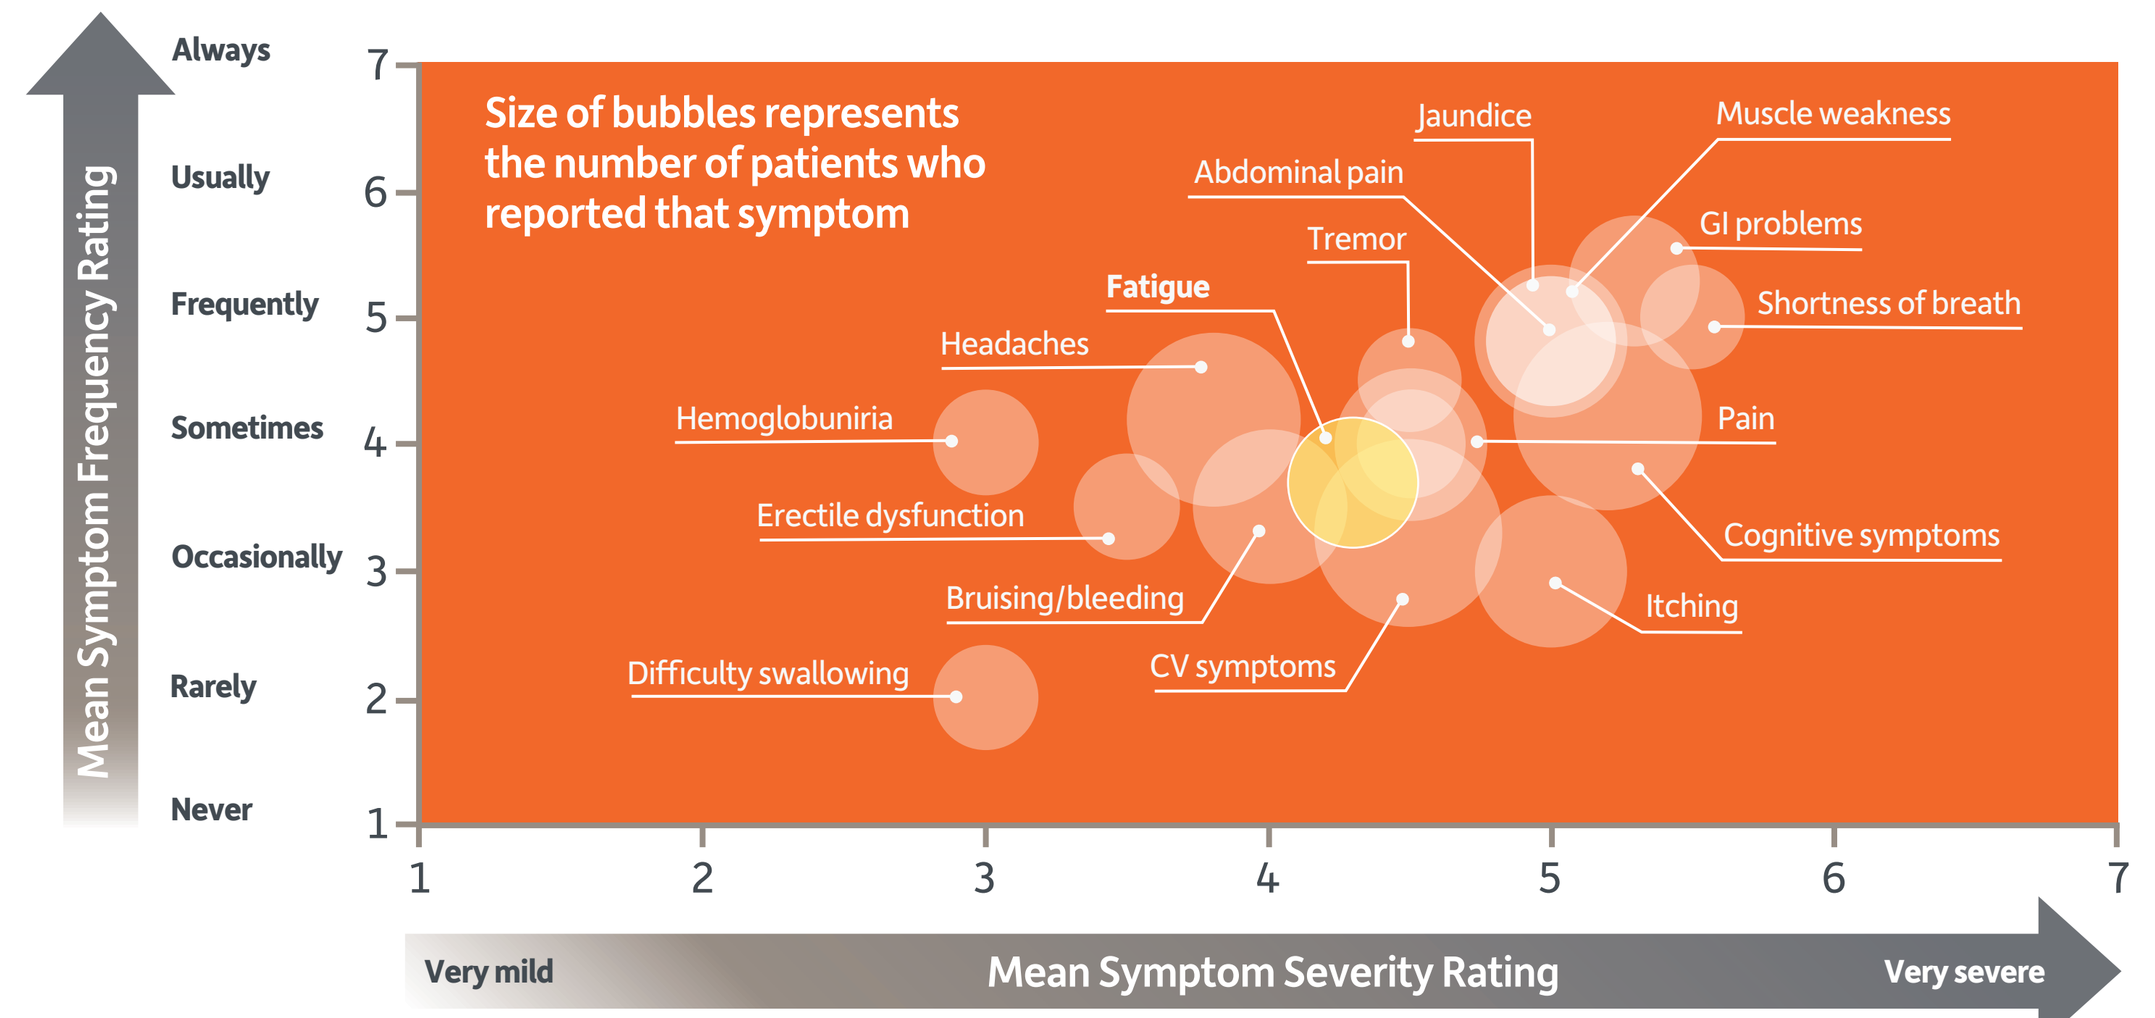
Always
Usually
Frequently
Sometimes
Occasionally
Rarely
Never
Mean Symptom Frequency Rating
Size of bubbles represents the number of patients who reported that symptom
Jaundice
Muscle weakness
Abdominal pain
GI problems
Tremor
Shortness of breath
Fatigue
Headaches
Hemoglobuniria
Pain
Erectile dysfunction
Cognitive symptoms
Bruising/bleeding
Itching
Difficulty swallowing
CV symptoms
Very mild
Mean Symptom Severity Rating
Very severe

Symptoms that decreased most in frequency (Change in mean rating)
Shortness of breath (-1.5); Fatigue (-1.3); Hemoglobuniria (-1.2); Bruising/bleeding (-1.0)
Symptoms that decreased most in severity (Change in mean rating)
Fatigue (-1.8); Shortness of breath (-1.5; Pain (-1.2); Tremor (-1.2)
CV, cardiovascular; GI, gastrointestinal.

Figure 3. Mean symptom frequency before and after switching from C5 inhibitors to pegcetacoplan (N=15)

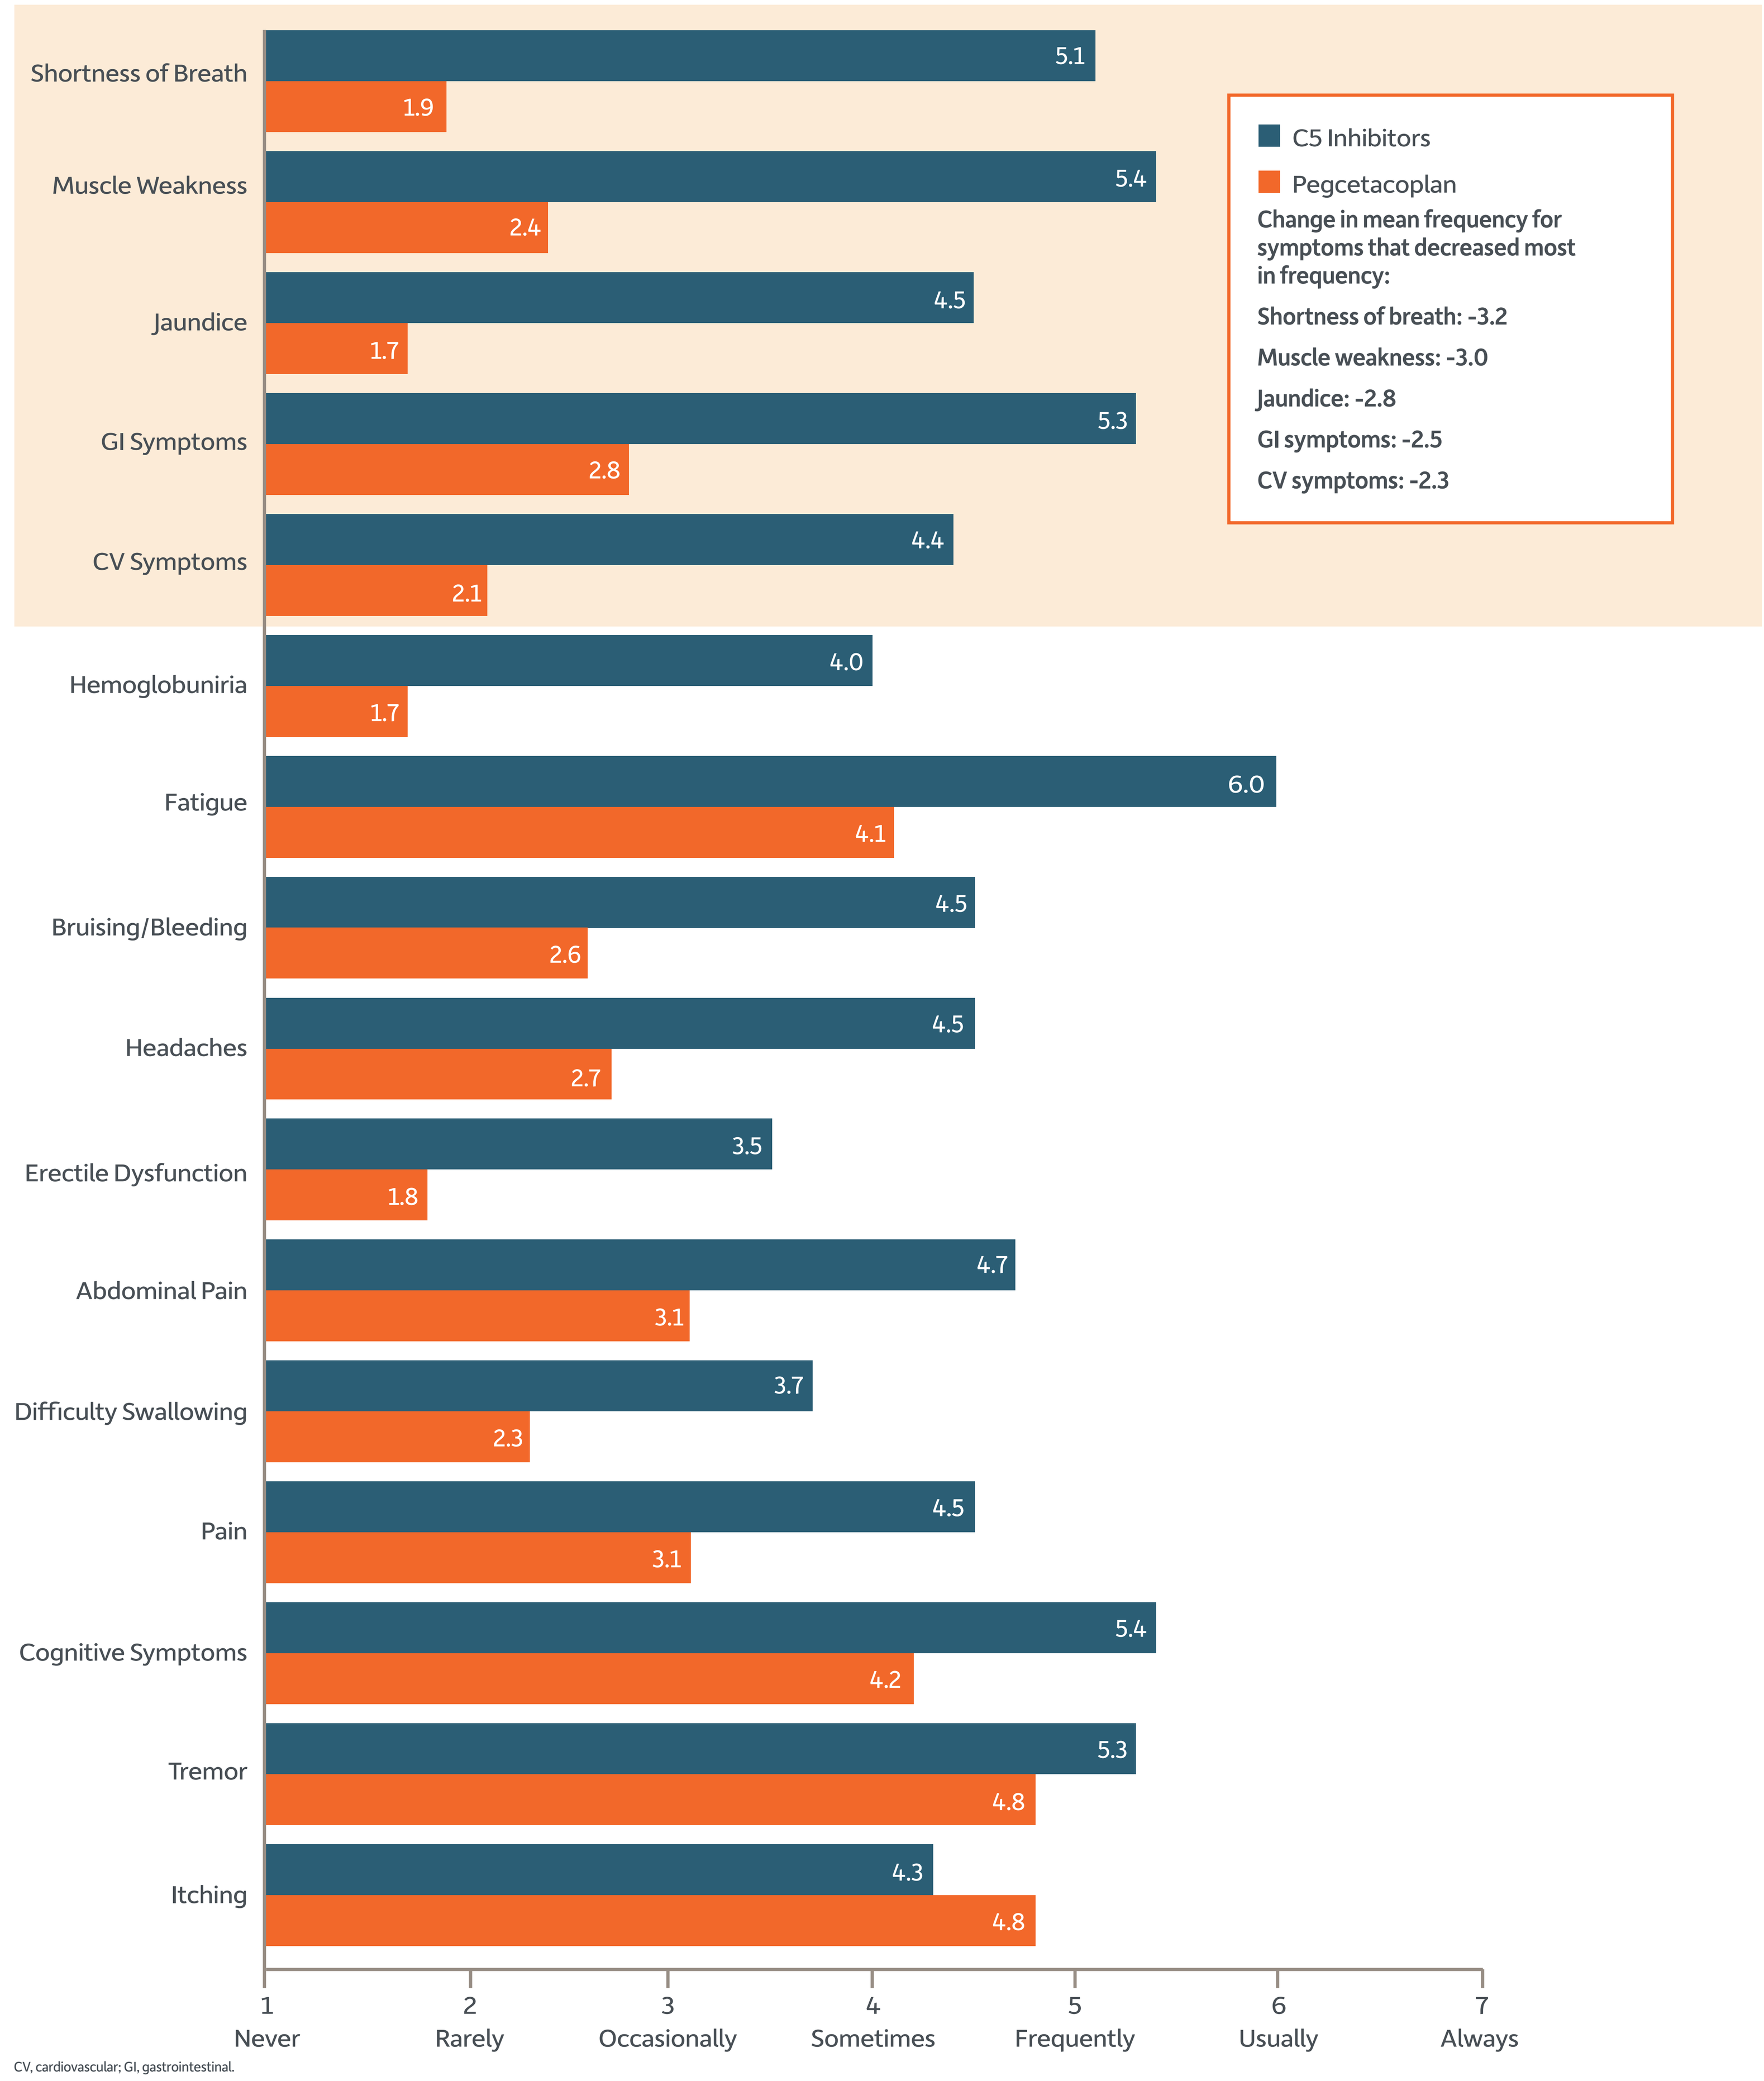
Shortness of Breath 5.1 1.9
Muscle Weakness 5.4 2.4
Jaundice 4.5 1.7
GI Symptoms 5.3 2.8
CV Symptoms 4.4 2.1
Hemoglobuniria 4.0 1.7
Fatigue 6.0 4.1
Bruising/Bleeding 4.5 2.6
Headaches 4.5 2.7
Erectile Dysfunction 3.5 1.8
Abdominal Pain 4.7 3.1
Difficulty Swallowing 3.7 2.3
Pain 4.5 3.1
Cognitive Symptoms 5.4 4.2
Tremor 5.3 4.8
Itching 4.3 4.8
1 Never
2 Rarely
3 Occasionally
4 Sometimes
5 Frequently
6 Usually
7 Always
C5 Inhibitors
Pegcetacoplan
Change in mean frequency for symptoms that decreased most in frequency:
Shortness of breath: -3.2
Muscle weakness: -3.0
Jaundice: -2.8
GI symptoms: -2.5
CV symptoms: -2.3

CV, cardiovascular; GI, gastrointestinal.

Figure 4. Mean symptom severity before and after switching from C5 inhibitors to pegcetacoplan (N=15)

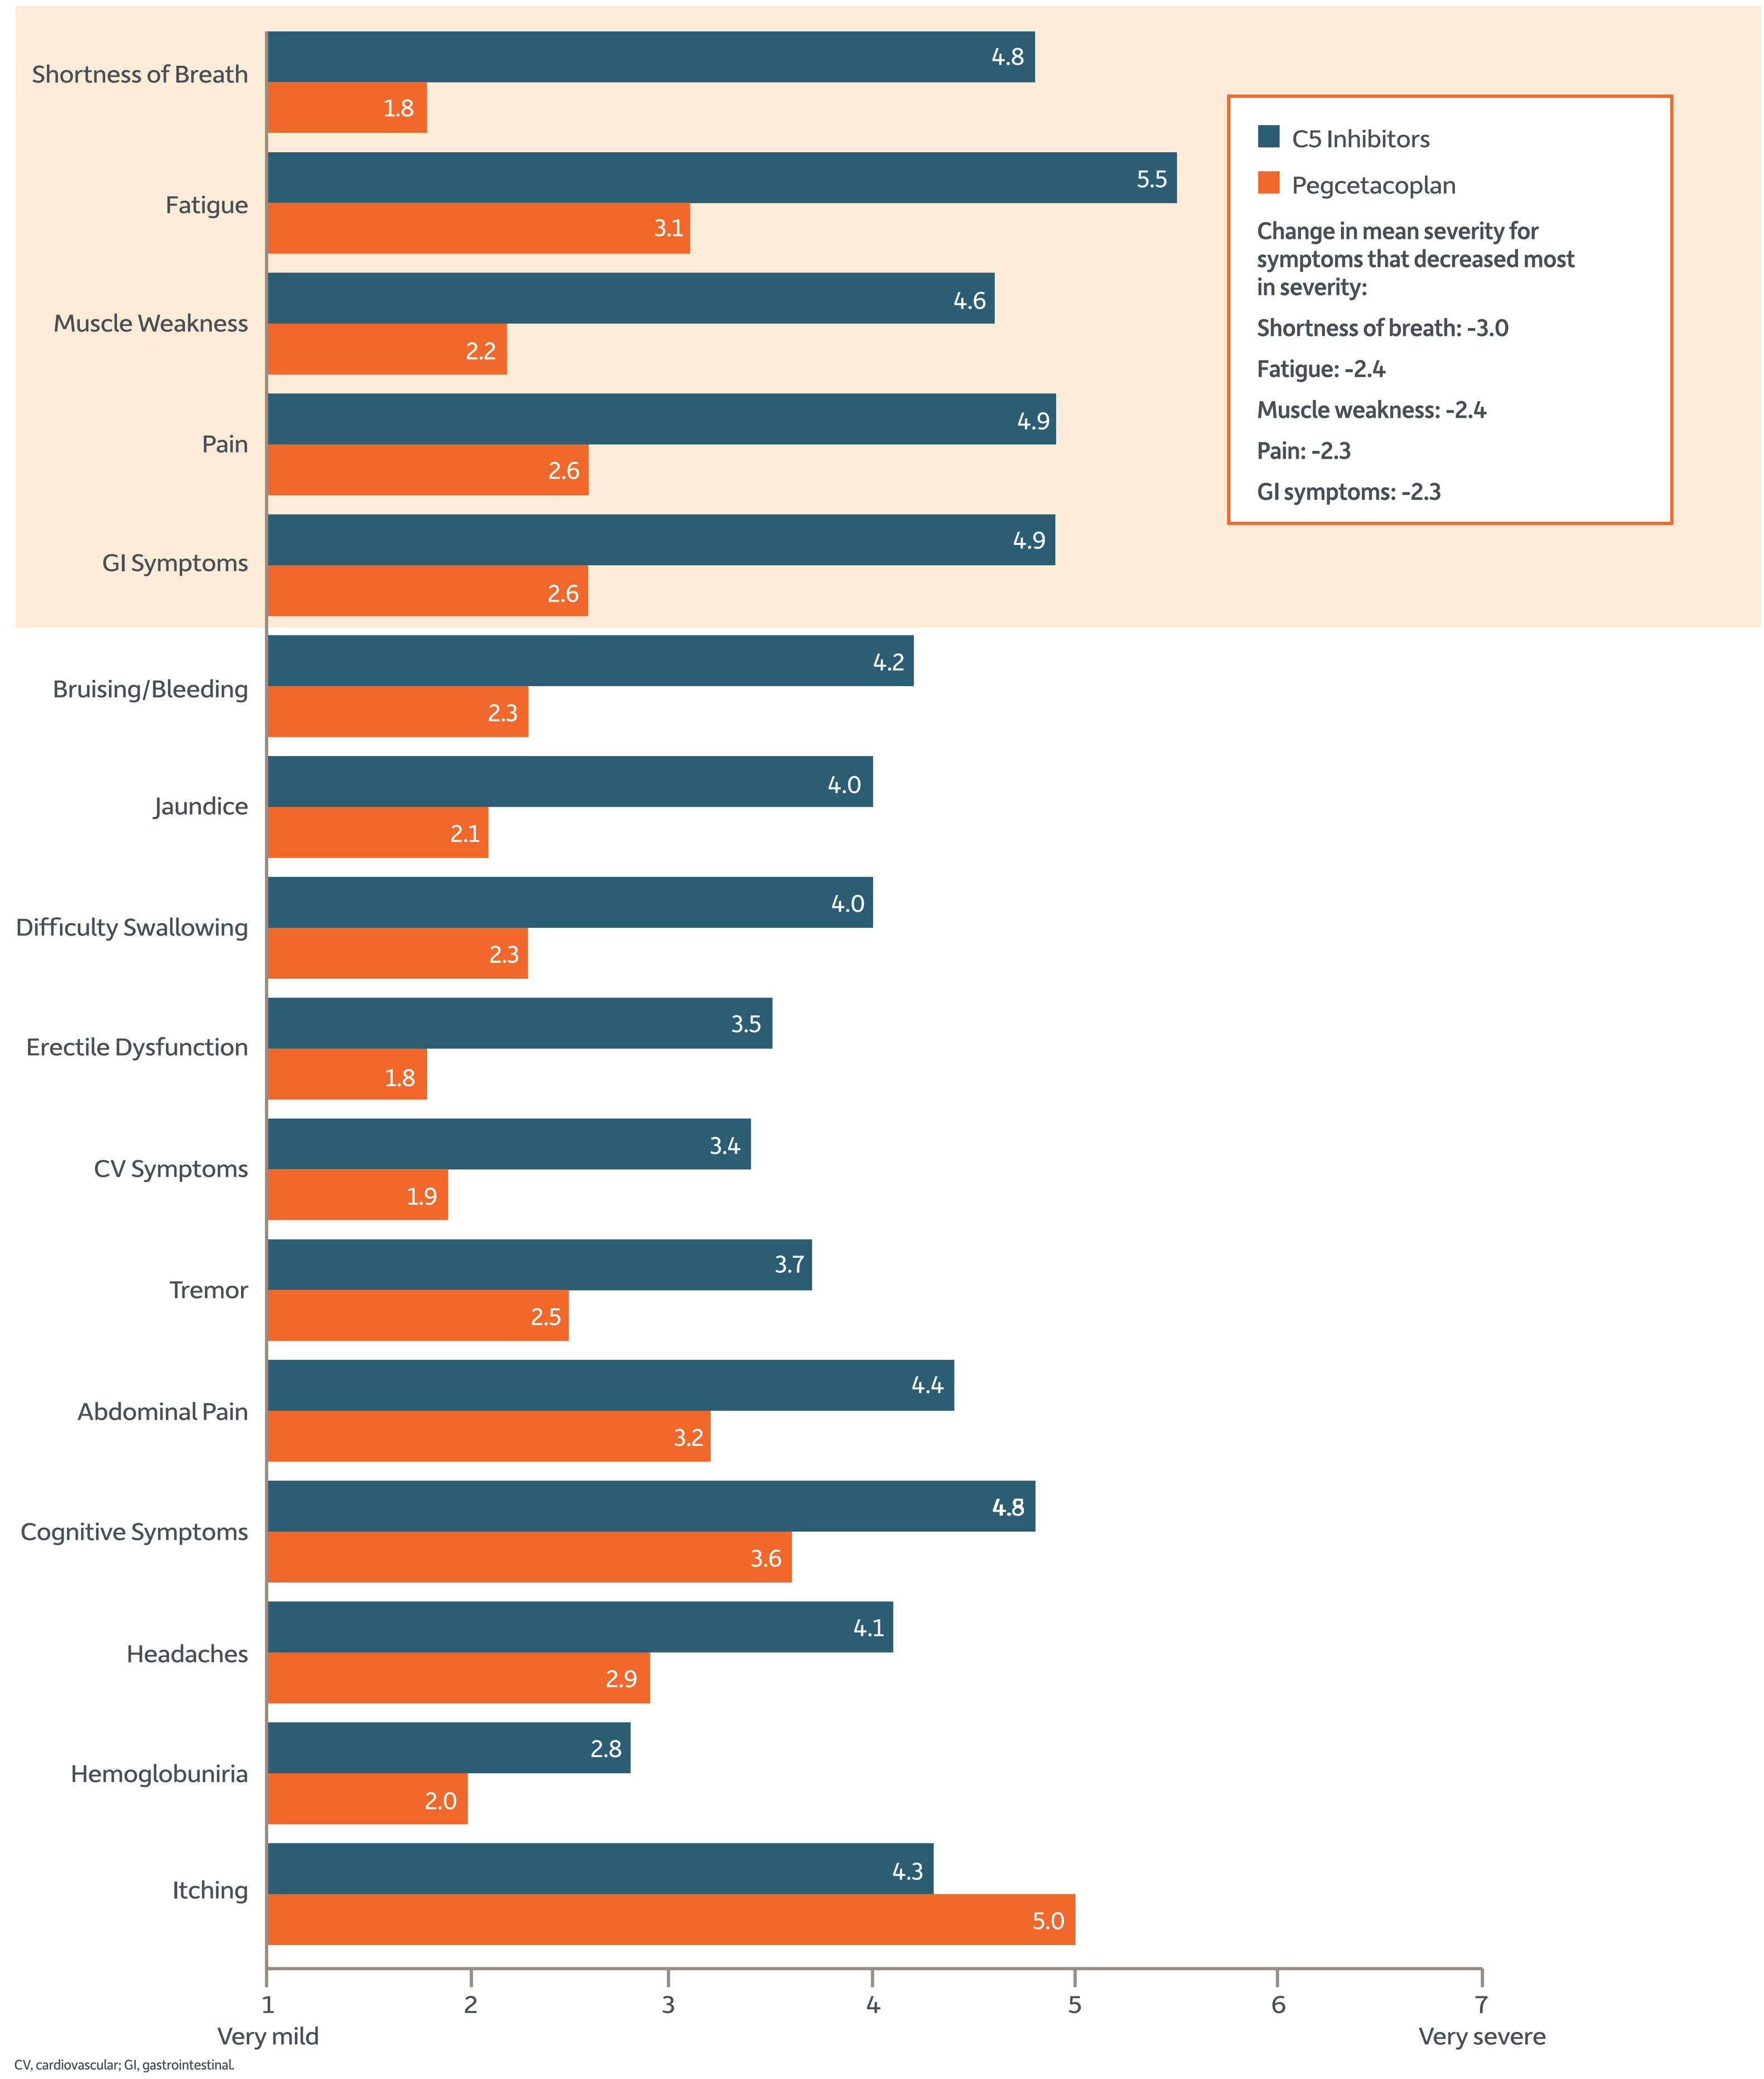
Shortness of Breath 4.8 1.8
Fatigue 5.5 3.1
Muscle Weakness 4.6 2.2
Pain 4.9 2.6
GI Symptoms 4.9 2.6
Bruising/Bleeding 4.2 2.3
Jaundice 4.0 2.1
Difficulty Swallowing 4.0 2.3
Erectile Dysfunction 3.5 1.8
CV Symptoms 3.4 1.9
Tremor 3.7 2.5
Abdominal Pain 4.4 3.2
Cognitive Symptoms 4.8 3.6
Headaches 4.1 2.9
Hemoglobuniria 2.8 2.0
Itching 4.3 5.0
1 Very mild
2
3
4
5
6
7 Very severe
C5 Inhibitors
Pegcetacoplan
Change in mean severity for symptoms that decreased most in severity:
Shortness of breath: -3.0
Fatigue: -2.4
Muscle weakness: -2.4
Pain: -2.3
GI symptoms: -2.3

CV, cardiovascular; GI, gastrointestinal.

Impact of switching to pegcetacoplan on PNH symptoms (continued)

- Patients reported relative reductions in the frequency of fatigue, cognitive symptoms, shortness of breath, muscle weakness, and GI symptoms after switching from C5 inhibitors to pegcetacoplan (**Figure 3**)
- A relative reduction in the severity of fatigue, cognitive symptoms, shortness of breath, muscle weakness, and GI symptoms was also observed after switching to pegcetacoplan (**Figure 4**)
- Select patients reported onset of new symptoms after switching, including shortness of breath (n=1/15) and muscle weakness (n=1/15); these were infrequent (1.0 and 2.0, respectively) and very mild (1.0 and 1.0, respectively)